8036

Ixazomib plus lenalidomide-dexamethasone (IRd) vs placebo-Rd in patients with relapsed/refractory multiple myeloma: China continuation of TOURMALINE-MM1

Jian Hou,[1] Jie Jin,[2] Yan Xu,[3] Depei Wu,[4] Xiaoyan Ke,[5] Zhou Daobin,[6] Jin Lu,[7] Xin Du,[8] Xiequn Chen,[9] Junmin Li,[10] Jing Liu,[11] Bingxia Wang,[12] Xiaoquan Zhang,[12] Hui Wang,[12] Helgi Van De Velde,[12] Paul G. Richardson,[13] Philippe Moreau[14]

[1]ChangZheng Hospital, The Second Military Medical University, Shanghai, China; [2]First Hospital Affiliated Zhe Jiang Medical University, Hangzou, China; [3]Institute of Hematology & Blood Diseases Hospital, Chinese Academy of Medical Science & Peking Union Medical College, Tianjin, China; [4]The First Affiliated Hospital of SooChow University, Jiangsu, China; [5]The Third Affiliated Hospital of Beijing Medical University, Beijing, China; [6]Peking Union Medical College Hospital, Beijing, China; [7]Peking University People's Hospital, Beijing, China; [8]Guangdong General Hospital/ Guangdong Academy of Medical Sciences, Guangzhou, China; [9]Xijing Hospital, The Fourth Military Medical University, Xi'an, China; [10]Ruijin Hospital, Shanghai Jiao Tong University, Shanghai, China; [11]The Third Hospital Xiang Ya Medical University, Changsha, China; [12]Millennium Pharmaceuticals, Inc., Cambridge, MA, USA a wholly owned subsidiary of Takeda Pharmaceutical Company Limited; [13]Dana-Farber Cancer Institute, Boston, MA, USA; [14]University Hospital Hôtel-Dieu, Nantes, France

Corresponding author contact information: houjian167@sohu.com

Background

- The introduction of novel therapies such as PIs and immunomodulatory compounds have significantly improved outcomes in MM.[1]
 - However, despite these improvements, the majority of patients with MM will ultimately relapse.[2]
- Data suggest that long-term therapy leads to better outcomes in MM, and treatment patterns are shifting towards extended treatment.[3]
 - However, the feasibility of long-term treatment with current regimens can be limited due to toxicities or the need for regular clinic attendance.[3]
- Ixazomib is the first oral PI to be studied in the clinic.
- In November 2015 the US FDA approved ixazomib in combination with lenalidomide and dexamethasone for the treatment of patients with MM who have received at least one prior therapy.[4]
- Approval was based on results from the global, randomized, double-blind, placebo-controlled, phase 3 TOURMALINE-MM1 study (NCT01564537) which investigated ixazomib plus lenalidomide/dexamethasone (IRd) vs placebo-Rd in 722 patients with RRMM following 1–3 prior therapies:[5]
 - IRd demonstrated a 35% improvement in PFS compared with placebo-Rd (HR 0.74, p=0.012), with limited additional toxicity.
- After completion of enrollment in the global TOURMALINE-MM1 study a continuation of this study was conducted in China as a separate regional expansion of the global study.

Objectives

- The aim of the China continuation study was to assess the efficacy and safety of IRd vs placebo-Rd in patients with RRMM in China.
- The primary endpoint was PFS.
- Secondary endpoints included OS, TTP, response rate, duration of response and safety.

Methods

Study design

- This was a double-blind, randomized, placebo-controlled study with an identical design to the global TOURMALINE-MM1 study[5] in terms of enrollment criteria, stratification factors, dosing regimen, dose modification guidelines, and study schedule (**Figure 1**).

Figure 1. Study design

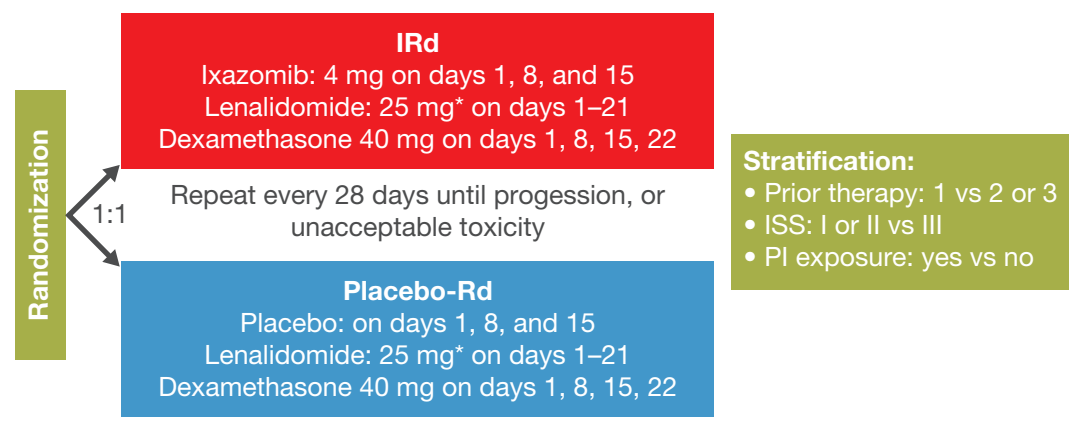
Randomization
1:1
IRd
Ixazomib: 4 mg on days 1, 8, and 15
Lenalidomide: 25 mg* on days 1–21
Dexamethasone 40 mg on days 1, 8, 15, 22
Repeat every 28 days until progession, or unacceptable toxicity
Placebo-Rd
Placebo: on days 1, 8, and 15
Lenalidomide: 25 mg* on days 1–21
Dexamethasone 40 mg on days 1, 8, 15, 22
Stratification:
• Prior therapy: 1 vs 2 or 3
• ISS: I or II vs III
• PI exposure: yes vs no

*10 mg for patients with creatinine clearance ≤60 or ≤50 mL/min, depending on local label/practice

Eligibility criteria

- Key inclusion criteria were:
 - Confirmed diagnosis of MM
 - Measurable disease by at least 1 of:
 - Serum protein electrophoresis
 - Urine protein electrophoresis
 - FLC assay
 - Received 1–3 prior treatments
 - Relapsed and/or refractory disease
 - Including primary refractory patients (i.e., patients refractory to all prior therapies)
 - Refractory = PD on treatment or within 60 days after last dose of therapy
 - Creatinine clearance ≥30 mL/min.
- Patients who were refractory to previous PI-based or lenalidomide-based treatment were excluded.

Assessments and Analyses

- Response and progression were assessed based on central laboratory determination of M-protein and FLC levels using IMWG 2011 criteria[6] and were evaluated by the same IRC as the global study.
- AEs were evaluated according to NCI CTCAE, Version 4.03.
- Patients were analyzed separately from the global study and were not included in the global TOURMALINE-MM1 intent-to-treat population.
 - Sample size was not based on a formal statistical hypothesis but intended to assess consistency in treatment effect with the global study.
 - A final analysis of PFS was to be completed either 18 months after the first patient enrolled in the China continuation or when a total of 60 PFS events were reached for patients in China, whichever came first.
 - At a data cut-off of 12 July 2015 the target number of PFS events had been observed; data presented here are based on this final analysis of PFS.

Results

Patients

- A total of 115 patients were randomized (1:1) and treated with IRd (n=57) or placebo-Rd (n=58).
 - Baseline characteristics are shown in **Table 1**.
- Compared with the global study, patients in the China continuation study had more advanced disease, which is consistent with the results of a large retrospective analysis of outcomes of Chinese patients with MM.[7]
 - At initial diagnosis, more patients had Durie-Salmon Stage IIIA myeloma (63% of Chinese patients vs 38% in the global population) and ISS stage III disease (37% vs 22%, respectively).
- In addition, compared with the global study, patients were more heavily pretreated (38% and 17% had received 2 or 3 prior lines of therapy vs 29% and 10%), more frequently had received prior thalidomide (84% vs 45%), more frequently had refractory MM (53% vs 11%), and more frequently had thalidomide refractory MM (63% vs 12%).

Efficacy

- At data cut-off, with a median follow-up of 8.0 and 7.8 months, respectively, PFS events had occurred in 30 patients (53%) in the IRd arm and 37 patients (64%) in the placebo-Rd arm.
- PFS was significantly improved with IRd vs placebo-Rd: median PFS 6.7 vs 4.0 months; HR 0.598; 95% CI 0.367–0.972; p=0.035 (**Figure 2**).
- The PFS benefit was seen across most prespecified subgroups including those with ISS stage I/II disease at screening, patients who had received 2/3 prior therapies, or prior PI/immunomodulatory compound therapy (**Figure 3**).
- TTP was also significantly improved with IRd vs placebo-Rd:
 - Median TTP was 7.3 vs 4.1 months; HR=0.583; p=0.032.
- Response rates were consistently higher and responses were of longer duration with IRd vs placebo-Rd (**Table 2**).

Table 1. Baseline characteristics

Baseline characteristic	IRd (N=57) n (%)	Placebo-Rd (N=58) n (%)	Total (N=115) n (%)
Age			
≤65	42 (74)	41 (71)	83 (72)
>65–75	14 (25)	14 (24)	28 (24)
>75	1 (2)	3 (5)	4 (3)
Sex: Male / Female	41 (72) / 16 (28)	38 (66) / 20 (34)	79 (69) / 36 (31)
Lines of prior therapy			
1	25 (44)	26 (45)	51 (44)
2	20 (35)	24 (41)	44 (38)
3	12 (21)	8 (14)	20 (17)
PI: exposed / naïve	34 (60) / 23 (40)	35 (60) / 23 (40)	69 (60) / 46 (40)
ISS Stage			
I	31 (54)	38 (66)	69 (60)
II	21 (37)	16 (28)	37 (32)
III	5 (9)	4 (7)	9 (8)
Disease status			
Relapsed*	15 (26)	13 (22)	28 (24)
Refractory†	28 (49)	33 (57)	61 (53)
Refractory and relapsed‡	14 (25)	12 (21)	26 (23)
Type of prior regimens			
Prior PI (all bortezomib)	34 (60)	35 (60)	69 (60)
Prior immunomodulatory therapy	52 (91)	47 (81)	99 (86)
Lenalidomide	2 (4)	7 (12)	9 (8)
Thalidomide	52 (91)	45 (78)	97 (84)
Thalidomide refractory	37 (65)	35 (60)	72 (63)
Corticosteroid	56 (98)	58 (100)	114 (99)
Dexamethasone	55 (96)	56 (97)	111 (97)
Prednisone	17 (30)	20 (34)	37 (32)
Melphalan	21 (37)	22 (38)	43 (37)
Prior stem cell transplant	8 (14)	12 (21)	20 (17)

*Patients who relapsed from ≥1 previous treatment (>60 days after the last dose of treatment) but were not refractory to any previous treatment.
†Patients who had disease progression on treatment or progression within 60 days after the last dose of ≥1 previous treatment but were not relapsed to any previous treatment.
‡Patients who relapsed from ≥1 previous treatment and were refractory to ≥1 previous treatment.

Figure 2. Kaplan-Meier plot of PFS (IRC assessment)

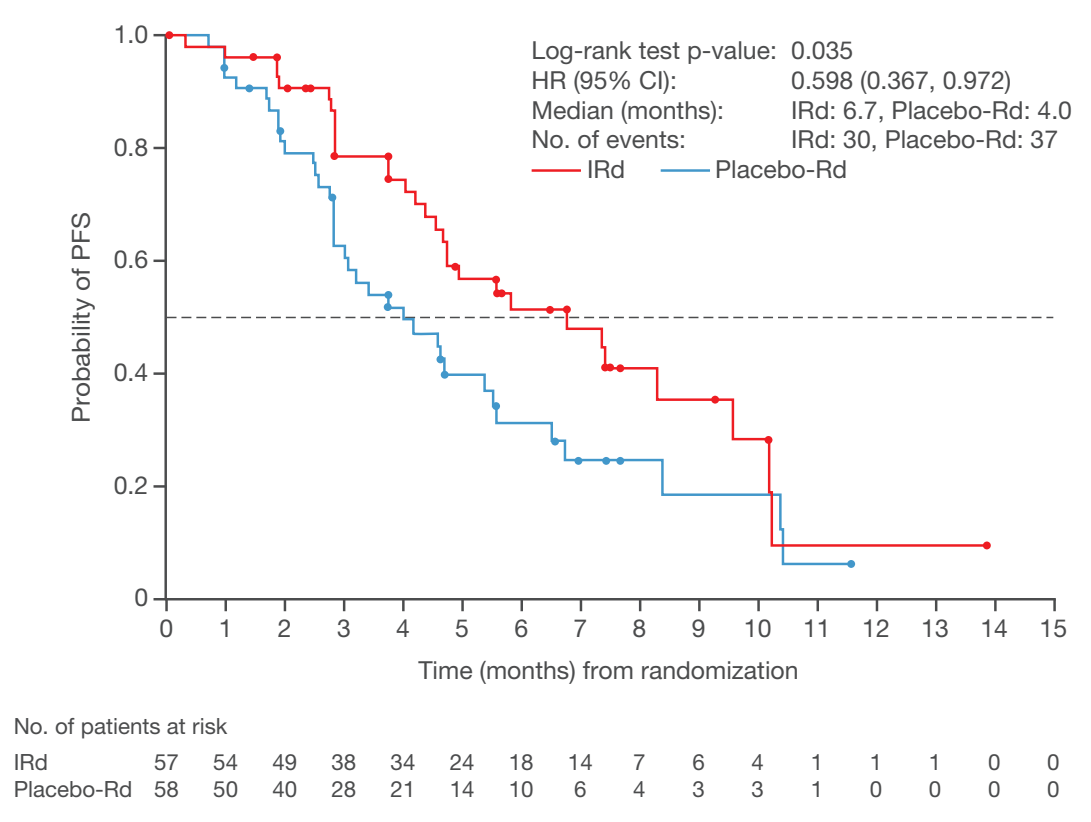
Log-rank test p-value: 0.035
HR (95% CI): 0.598 (0.367, 0.972)
Median (months): IRd: 6.7, Placebo-Rd: 4.0
No. of events: IRd: 30, Placebo-Rd: 37
IRd
Placebo-Rd
Probability of PFS
1.0
0.8
0.6
0.4
0.2
0
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15
Time (months) from randomization
No. of patients at risk
IRd 57 54 49 38 34 24 18 14 7 6 4 1 1 1 0 0
Placebo-Rd 58 50 40 28 21 14 10 6 4 3 3 1 0 0 0 0

Figure 3. Forest plot of PFS with IRd vs placebo-Rd in prespecified patient subgroups in the China continuation population

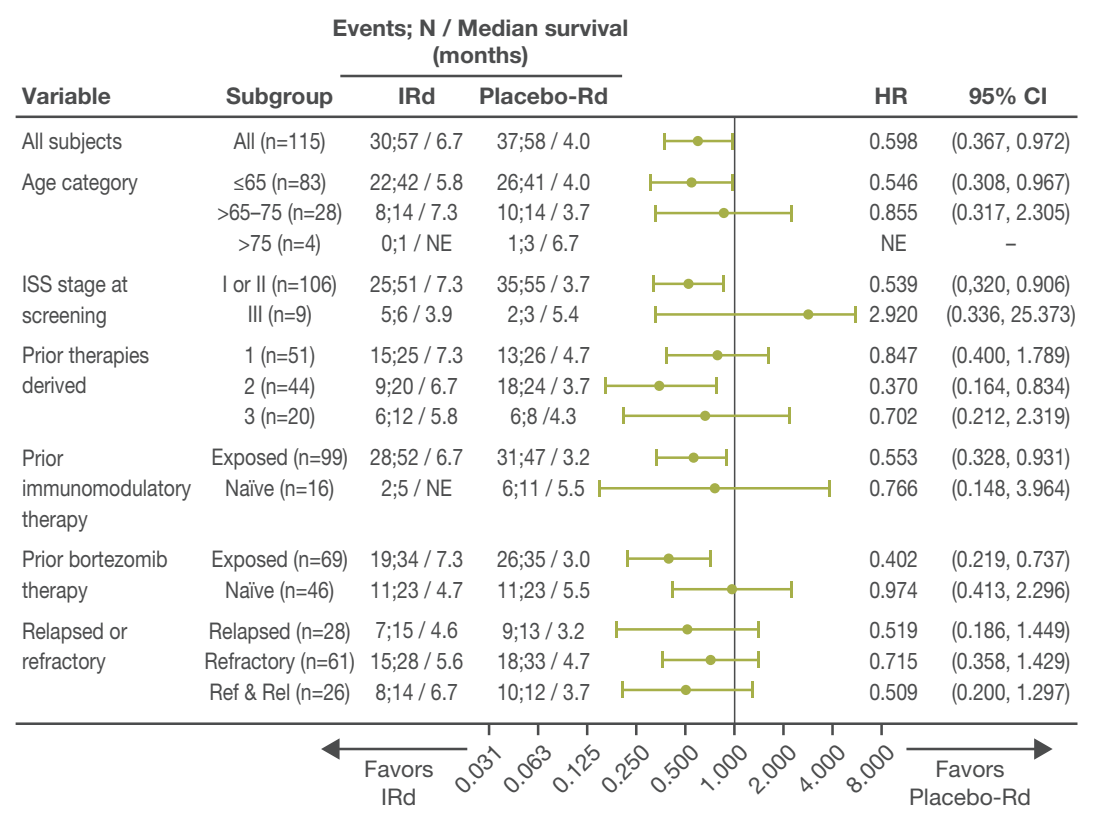
Events; N / Median survival (months)
Variable | Subgroup | IRd | Placebo-Rd | HR | 95% CI
All subjects | All (n=115) | 30;57 / 6.7 | 37;58 / 4.0 | 0.598 | (0.367, 0.972)
Age category | ≤65 (n=83) | 22;42 / 5.8 | 26;41 / 4.0 | 0.546 | (0.308, 0.967)
>65–75 (n=28) | 8;14 / 7.3 | 10;14 / 3.7 | 0.855 | (0.317, 2.305)
>75 (n=4) | 0;1 / NE | 1;3 / 6.7 | NE | –
ISS stage at screening | I or II (n=106) | 25;51 / 7.3 | 35;55 / 3.7 | 0.539 | (0,320, 0.906)
III (n=9) | 5;6 / 3.9 | 2;3 / 5.4 | 2.920 | (0.336, 25.373)
Prior therapies derived | 1 (n=51) | 15;25 / 7.3 | 13;26 / 4.7 | 0.847 | (0.400, 1.789)
2 (n=44) | 9;20 / 6.7 | 18;24 / 3.7 | 0.370 | (0.164, 0.834)
3 (n=20) | 6;12 / 5.8 | 6;8 /4.3 | 0.702 | (0.212, 2.319)
Prior immunomodulatory therapy | Exposed (n=99) | 28;52 / 6.7 | 31;47 / 3.2 | 0.553 | (0.328, 0.931)
Naïve (n=16) | 2;5 / NE | 6;11 / 5.5 | 0.766 | (0.148, 3.964)
Prior bortezomib therapy | Exposed (n=69) | 19;34 / 7.3 | 26;35 / 3.0 | 0.402 | (0.219, 0.737)
Naïve (n=46) | 11;23 / 4.7 | 11;23 / 5.5 | 0.974 | (0.413, 2.296)
Relapsed or refractory | Relapsed (n=28) | 7;15 / 4.6 | 9;13 / 3.2 | 0.519 | (0.186, 1.449)
Refractory (n=61) | 15;28 / 5.6 | 18;33 / 4.7 | 0.715 | (0.358, 1.429)
Ref & Rel (n=26) | 8;14 / 6.7 | 10;12 / 3.7 | 0.509 | (0.200, 1.297)
Favors IRd
0.031 0.063 0.125 0.250 0.500 1.000 2.000 4.000 8.000
Favors Placebo-Rd

Table 2. Response to treatment

Best confirmed response	IRd (N=57) n (%)	Placebo-Rd (N=58) n (%)	p-value*
CR	3 (5)	0	0.078
PR	29 (51)	18 (31)	
VGPR	11 (19)	7 (12)	
ORR (CR + VGPR + PR)	32 (56)	18 (31)	0.007
CR + VGPR rate	14 (25)	7 (12)	0.084
SD	17 (30)	17 (29)	
PD	6 (11)	15 (26)	
NE	2 (4)	8 (14)	

*p-value is from an unstratified CMH test.

- Among responders, the median duration of response was 7.4 months with IRd vs 5.6 months with placebo-Rd.
- OS data are not yet mature:
 - 6 (11%) patients treated with IRd and 16 (28%) patients treated with placebo-Rd have died
 - The study remains blinded and is ongoing with a final analysis for mature OS data planned.

Safety

- At data cut-off, patients in the IRd and placebo-Rd arms had received a median of 7 and 5 treatment cycles, respectively, and 59% and 41% of patients remained on treatment.
 - The main reasons for treatment discontinuation in the IRd and placebo-Rd arms were progressive disease (33% and 41%) and AEs (5% and 12%).
- The median relative dose intensity (RDI; amount of drug received as a proportion of the amount of drug expected/prescribed) of all drugs was high, and consistent with the global study[5] (**Table 3**).

Table 3. Median RDI for study drugs by treatment arm

Median RDI (%)	IRd (n=57)	Placebo-Rd (n=58)
Blinded study drug (ixazomib/placebo)	100	100
Lenalidomide	96.0	99.8
Dexamethasone	95.5	97.1

- The overall safety profile was similar in both treatment groups (**Table 4**).

Table 4. Summary of treatment-emergent AEs

Preferred term	IRd (n=57) n (%)	Placebo-Rd (n=58) n (%)	Total (N=115) n (%)
Any AE	57 (100)	55 (95)	112 (97)
Grade ≥3 AE	32 (56)	36 (62)	68 (59)
Drug-related AE	53 (93)	55 (95)	108 (94)
Drug-related grade ≥3 AE	29 (51)	32 (55)	61 (53)
SAE	13 (23)	15 (26)	28 (24)
Drug-related SAE	9 (16)	5 (9)	14 (12)
AEs resulting in any study regimen drug dose modification*	19 (33)	22 (38)	41 (36)
AEs resulting in discontinuation of the full study regimen	3 (5)	7 (12)	10 (9)
AEs resulting in any study regimen drug dose reduction	9 (16)	6 (10)	15 (13)
AEs resulting in any study regimen drug dose discontinuation	4 (7)	7 (12)	11 (10)
On-study deaths†	2 (4)	3 (5)	5 (4)

*Includes dose reduction, dose increase, dose delay, and dose discontinuation; †Defined as deaths during treatment or within 30 days after the last dose of any study drug.

- Common AEs and grade ≥3 AEs with IRd vs placebo-Rd are shown in **Table 5**.

Table 5. Treatment-emergent AEs of clinical interest

Pooled preferred terms	IRd (n=57) n (%) Overall	IRd Grade ≥3	Placebo-Rd (n=58) n (%) Overall	Placebo-Rd Grade ≥3
Thrombocytopenia*	38 (67)	13 (23)	32 (55)	8 (13)
Neutropenia†	26 (46)	13 (23)	26 (45)	11 (19)
Anemia‡	14 (25)	7 (12)	27 (47)	15 (26)
Pneumonia	12 (21)	9 (16)	10 (17)	6 (10)
Rash	10 (18)	0	11 (19)	0
Hepatic events	10 (18)	3 (5)	5 (9)	0
Diarrhea	9 (16)	1 (2)	2 (3)	0
Nausea	5 (9)	0	2 (3)	0
Vomiting	5 (9)	0	1 (2)	0
Peripheral neuropathies	4 (7)	0	4 (7)	0
Cardiac arrhythmias	3 (5)	0	1 (2)	0
Acute renal failure	2 (4)	1 (2)	5 (9)	3 (5)
Heart failure	1 (2)	1 (2)	1 (2)	1 (2)
Hypotension	1 (2)	0	0	0
New primary malignancy	0	0	1 (2)	1 (2)

*Thrombocytopenia and platelet count decreased; †Neutropenia and neutrophil count decreased; ‡Anemia and hemoglobin decreased.

- Ixazomib was associated with a decreased incidence of the possibly myeloma-related events of anemia and renal insufficiency:
 - The rate of grade ≥3 anemia was lower with IRd vs placebo-Rd (12% vs 26%), as was the use of RBC transfusions (9% vs 21%)
 - The incidence of AEs associated with renal impairment (as defined by acute renal failure) was lower with IRd vs placebo-Rd (all grade: 4% vs 9%; grade ≥3: 2% vs 5%, respectively)
 - Improvement in renal function reversal, defined as a shift from baseline creatinine clearance <50 mL/min to a post-baseline value >60 mL/min, was noted for a higher percentage of patients receiving IRd (2 out of 2 patients with creatinine clearance <50 mL/min at baseline; 100%) vs placebo-Rd (0 out of 5 patients with creatinine clearance <50 mL/min at baseline; 0%).
- Patients in the China continuation study reported a higher frequency of hematologic AEs in comparison to the global study.[5]
 - However, this was not reflected in the central laboratory results, which indicated a similar high grade shift of platelet and neutrophil counts in both populations; suggesting that the higher rates of hematologic AEs may be influenced by differences in AE reporting practices between China and global physicians.
- There was no evidence of cardiac or renal toxicity with the addition of ixazomib, and no incidences of venous thromboembolism or new primary malignancies.
- Based on an observed higher incidence of herpes zoster reactivation in the IRd treatment group (18%) vs placebo-Rd (0%), the IDMC recommended an antiviral prophylaxis requirement for the entire study population still on treatment; the China continuation protocol was amended accordingly.

Conclusions

- In Chinese patients with RRMM, IRd was associated with a significant improvement in PFS in ITT analysis.
 - A clear treatment benefit with IRd was additionally observed for the secondary efficacy endpoints of TTP, response rate, and duration of response.
 - The combination of IRd had limited additional toxicity over Rd alone.
- The data in this distinct Chinese population further support the conclusions of treatment benefit of IRd vs placebo-Rd observed in the global TOURMALINE-MM1 study.[5]
 - The magnitude of benefit with IRd vs placebo-Rd was similar to the global study.
 - Although the actual outcomes were different due to the differences in the patient population,[6] such as the extent of pretreatment, proportion of prior thalidomide therapy, and proportion of refractory and thalidomide-refractory patients.
 - Consistent with the global study, there was limited additional toxicity with IRd vs placebo-Rd.
- The study is ongoing in a blinded manner and survival and safety will continue to be monitored until the final analysis for OS.

References

1. Kumar S, et al. Leukemia 2014;28:1122–8.
2. Borrello I. Leuk Res 2012;36(Suppl. 1):S3–12.
3. Ludwig H, Sonneveld P. Leuk Res 2012;36(Suppl. 1):S27–34.
4. Millennium Pharmaceuticals. NINLARO® (ixazomib) capsules, for oral use. Prescribing information. November 2015. (http://www.ninlaro.com/downloads/prescribing-information.pdf)
5. Moreau P, et al. N Engl J Med 2016;374:1621–34.
6. Rajkumar S, et al. Blood 2011;117:4691–5.
7. Lu J, et al. Blood Cancer J 2014;4:e239.

Abbreviations

AE, adverse events; CI, confidence interval; CMH, Cochran-Mantel-Haenszel; CR, complete response; FDA, Food and Drug Agency; FLC, free light chain; HR, hazard ratio; IDMC, Independent Data Monitoring Committee; IMWG, International Myeloma Working Group; IRC, independent review committee; IRd, ixazomib plus lenalidomide-dexamethasone; ISS, International Staging System; ITT, intent-to-treat; MM, multiple myeloma; NCI CTCAE; National Cancer Institute Common Terminology Criteria for Adverse Events; NE, not evaluable; ORR, overall response rate; OS, overall survival; PD, progressive disease; PFS, progression-free survival; PI, proteasome inhibitor; PN, peripheral neuropathy; PR, partial response; RBC, red blood cells; Rd, lenalidomide-dexamethasone; RDI, relative dose intensity; RRMM, relapsed/refractory multiple myeloma; SAE, serious adverse events; VGPR, very good partial response; TTP, time to progression.

Acknowledgments

The authors would like to thank all patients and their families, and all investigators for their valuable contributions to the study. The authors acknowledge Helen Wilkinson of FireKite, an Ashfield company, part of UDG Healthcare plc, for writing assistance during the development of this poster, which was funded by Millennium Pharmaceuticals Inc., in compliance with Good Publication Practice 3 ethical guidelines (Battisti WP, et al. Ann Intern Med 2015;163:461–4).

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Poster presented at the American Society of Clinical Oncology (ASCO) Annual Meeting, Chicago, IL, USA, June 3–7, 2016